Outcomes in Patients Receiving Oral Octreotide Capsules Related to Time From Last SRL Injection: Analysis From the MPOWERED Phase 3 Study

This content is intended for healthcare professionals only

Macut D,[1] Fleseriu M,[2] Dreval A,[3] Pokramovich YG,[3] Bondar I,[4] Isaeva E,[5] Molitch ME,[6] Leonova N,[7] Haviv A,[8] Biermasz NR,[9] Melmed S,[10] Raverot G,[11] Strasburger CJ[12]

[1]Faculty of Medicine, University of Belgrade, Belgrade, Serbia; [2]Oregon Health & Science University, Portland, OR, USA; [3]M.F. Vladimirsky Moscow Regional Research Clinical Institute, Moscow, Russia; [4]Novosibirsk State Medical University, Novosibirsk, Russia; [5]Interregional Clinical Diagnostic Center, Kazan, Russia; [6]Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [7]Antrium Multidisciplinary Medical Clinic, Barnaul, Altai Region, Russia; [8]Amryt Pharmaceuticals DAC, Dublin, Ireland; [9]Leiden University Medical Center, Leiden, Netherlands; [10]Cedars–Sinai Medical Center, Los Angeles, CA, USA; [11]Hospices Civils de Lyon, Bron, France; [12]Charite–Universitätsmedizin, Campus Mitte, Berlin, Germany

Introduction

- Recent updates to acromegaly treatment guidelines recommend that oral octreotide capsules (OOC) should be initiated circa the time of the next scheduled injectable somatostatin receptor ligand (iSRL) dose, as was performed in the OPTIMAL study[1,2]
- In the MPOWERED phase 3 trial (NCT02685709), OOC could be initiated any time after the last injection up until the time of the next schedule injection +3 days[3]
- The impact of earlier initiation of OOC on safety and efficacy has not been previously investigated
- The objective of this analysis is to describe clinical and safety outcomes based on the time since the last iSRL dose and initiation of OOC during the 26-week Run-in phase of MPOWERED

Study Design

Inclusion criteria

- On a stable dose of iSRL for ≥4 months
- Biochemical response (IGF–I <1.3 × ULN and integrated GH <2.5 ng/mL) at screening

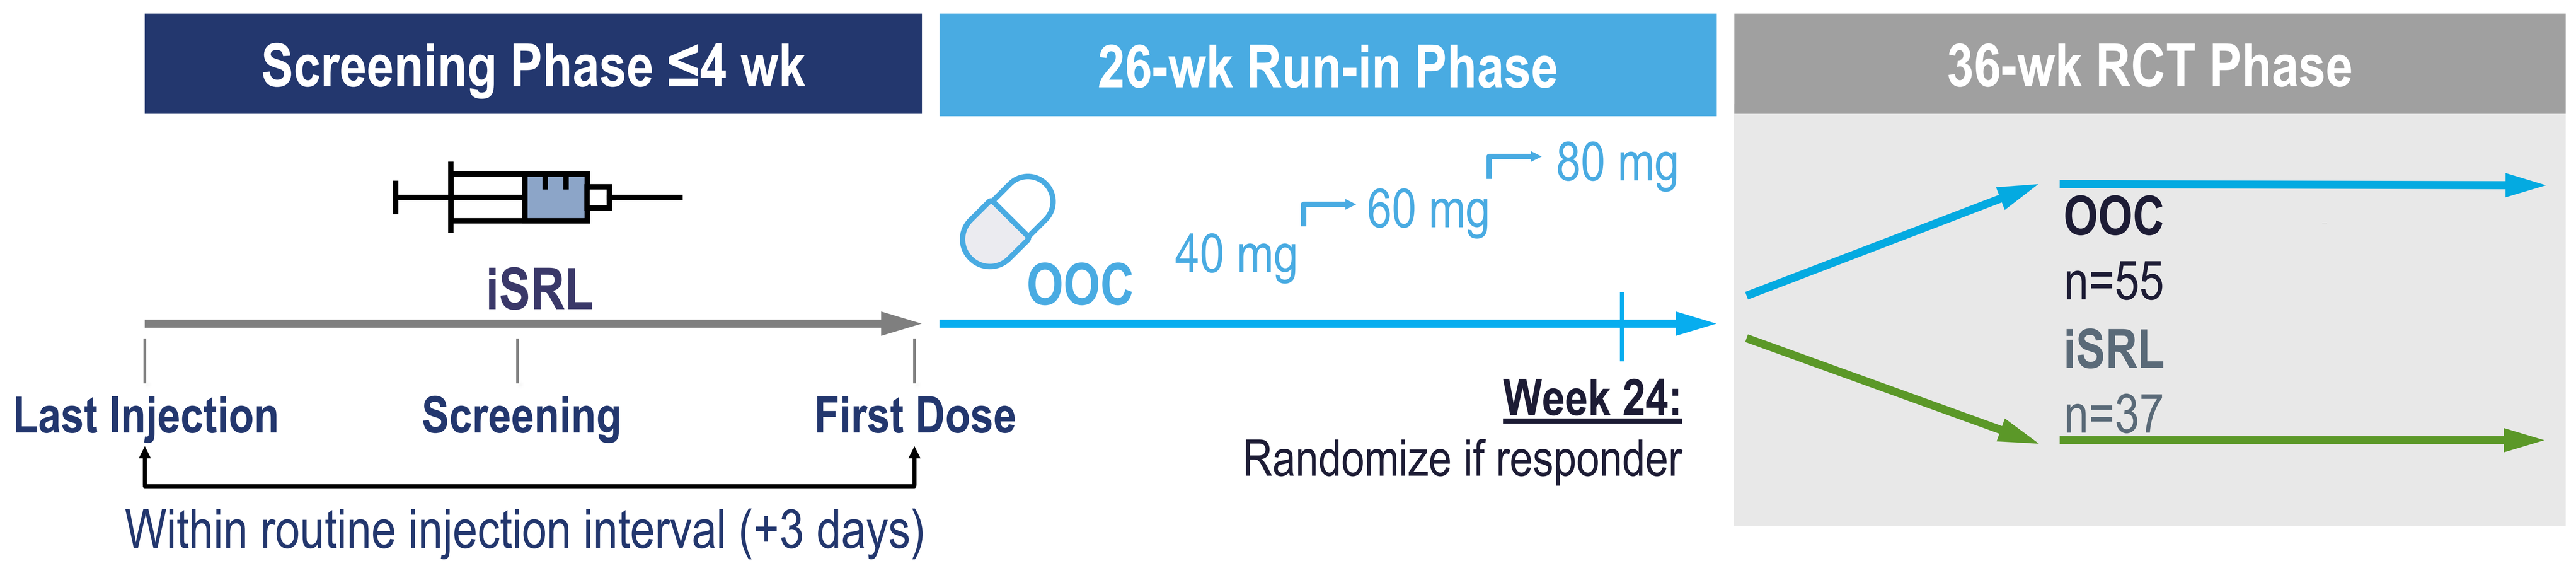
Screening Phase ≤4 wk
26-wk Run-in Phase
36-wk RCT Phase
iSRL
Last Injection
Screening
First Dose
Within routine injection interval (+3 days)
OOC
40 mg
60 mg
80 mg
Week 24:
Randomize if responder
OOC
n=55
iSRL
n=37

Populations Assessed		
Time since last iSRL dose	<3 wk (n=40)	≥3 wk (n=105)

GH, growth hormone; IGF-I, insulin-like growth factor I; iSRL, injectable somatostatin receptor ligand; OOC, oral octreotide capsules; RCT, randomized controlled treatment; SRL, somatostatin receptor ligand; ULN, upper limit of normal; wk, week.

Results

Time since last iSRL dose did not impact final OOC dose in the Run-in phase

Final OOC Dose in Run-in Phase

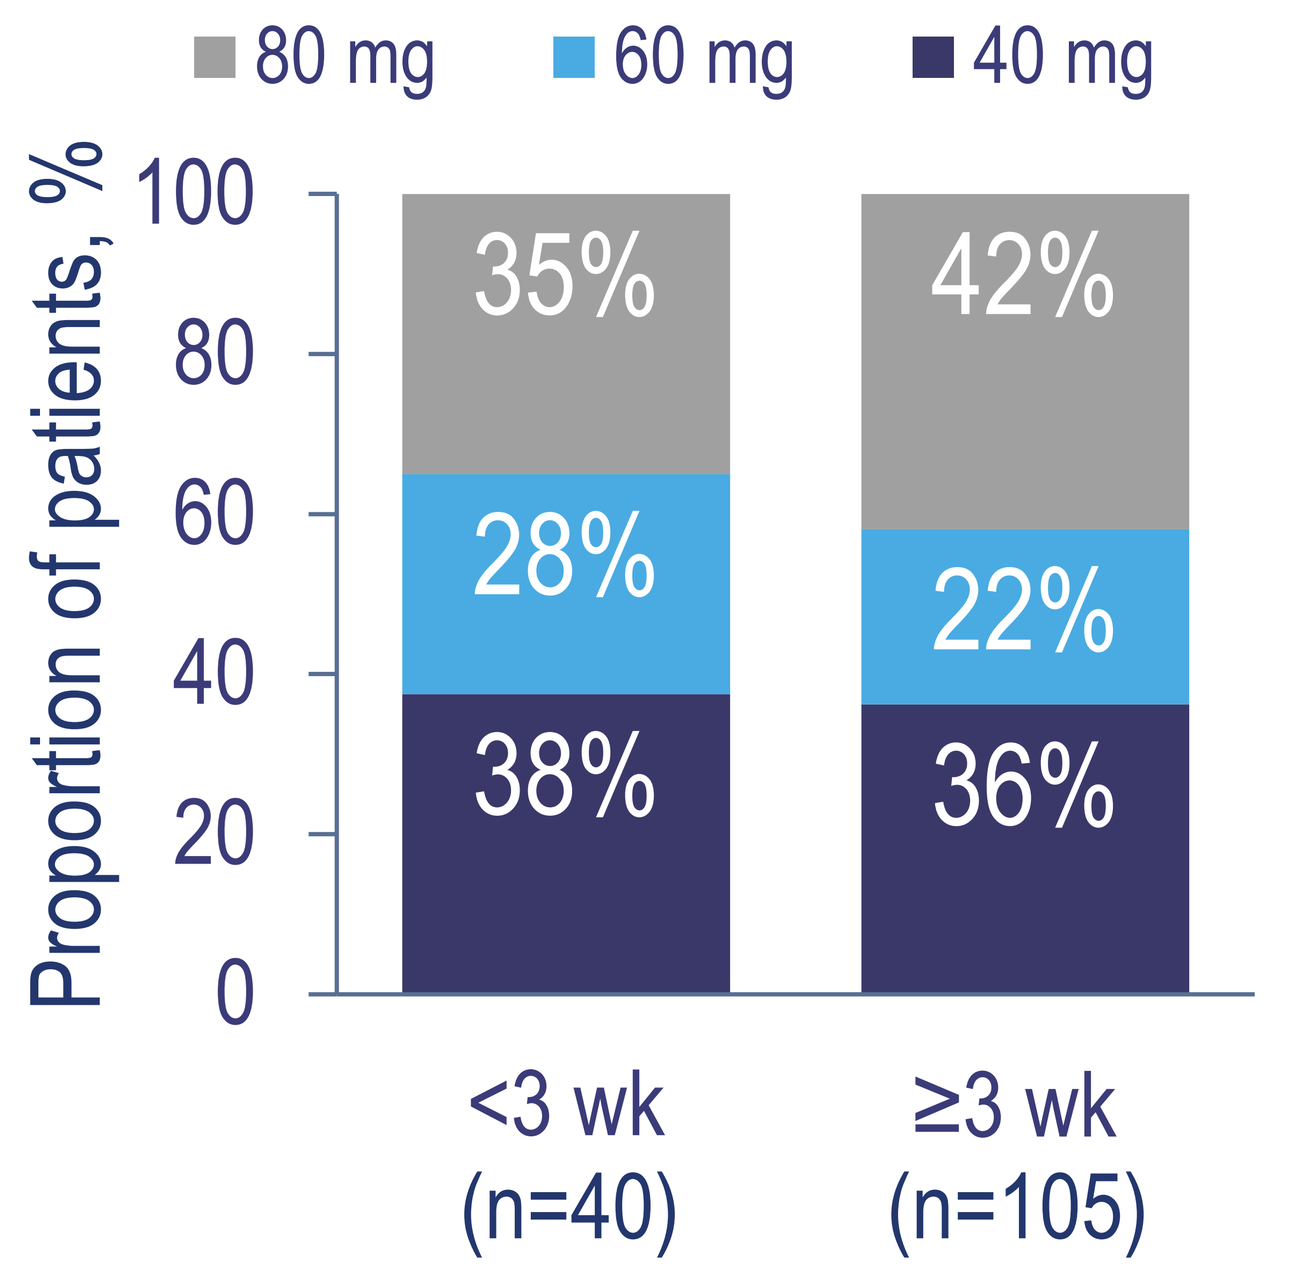
80 mg
60 mg
40 mg
Proportion of patients, %
100
80
60
40
20
0
35%
28%
38%
42%
22%
36%
<3 wk
(n=40)
≥3 wk
(n=105)
Time since last iSRL dose

Time since last iSRL dose

	<3 wk (n=40)	≥3 wk (n=105)
Completed Run-in phase, n (%)	35 (87.5)	81 (77.1)
Reasons for discontinuation, n (%)		
AE	1 (2.5)	13 (12.4)
Treatment failure	2 (5.0)	5 (4.8)
Withdrawal by subject	2 (5.0)	4 (3.8)
Protocol deviation	0	1 (1.0)
Investigator request	0	1 (1.0)

AE, adverse event.

IGF-I levels and maintenance of biochemical response (IGF-I <1.3 × ULN) were comparable during the Run-in phase, regardless of timing of OOC initiation

Maintenance of Biochemical Response

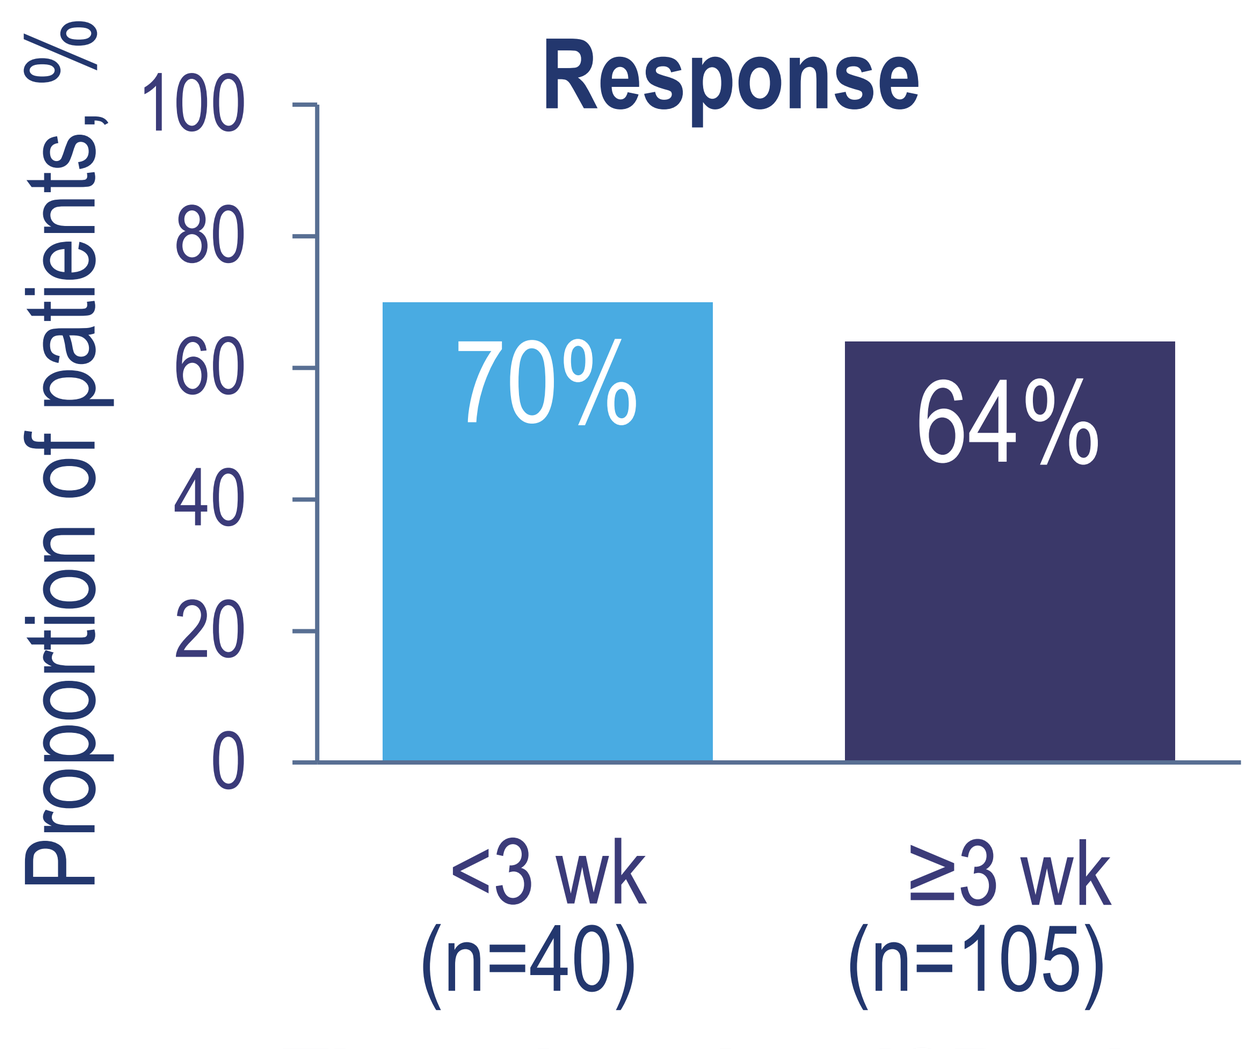
Proportion of patients, %
100
80
60
40
20
0
70%
64%
<3 wk
(n=40)
≥3 wk
(n=105)
Time since last iSRL dose

- Biochemical response was maintained in similar proportions in each group
- When evaluating the population of biochemical responders at the end of Run-in, mean IGF-I values remained within the normal limit at week 4 and week 26 of the Run-in in both groups

IGF-I Throughout Run-in

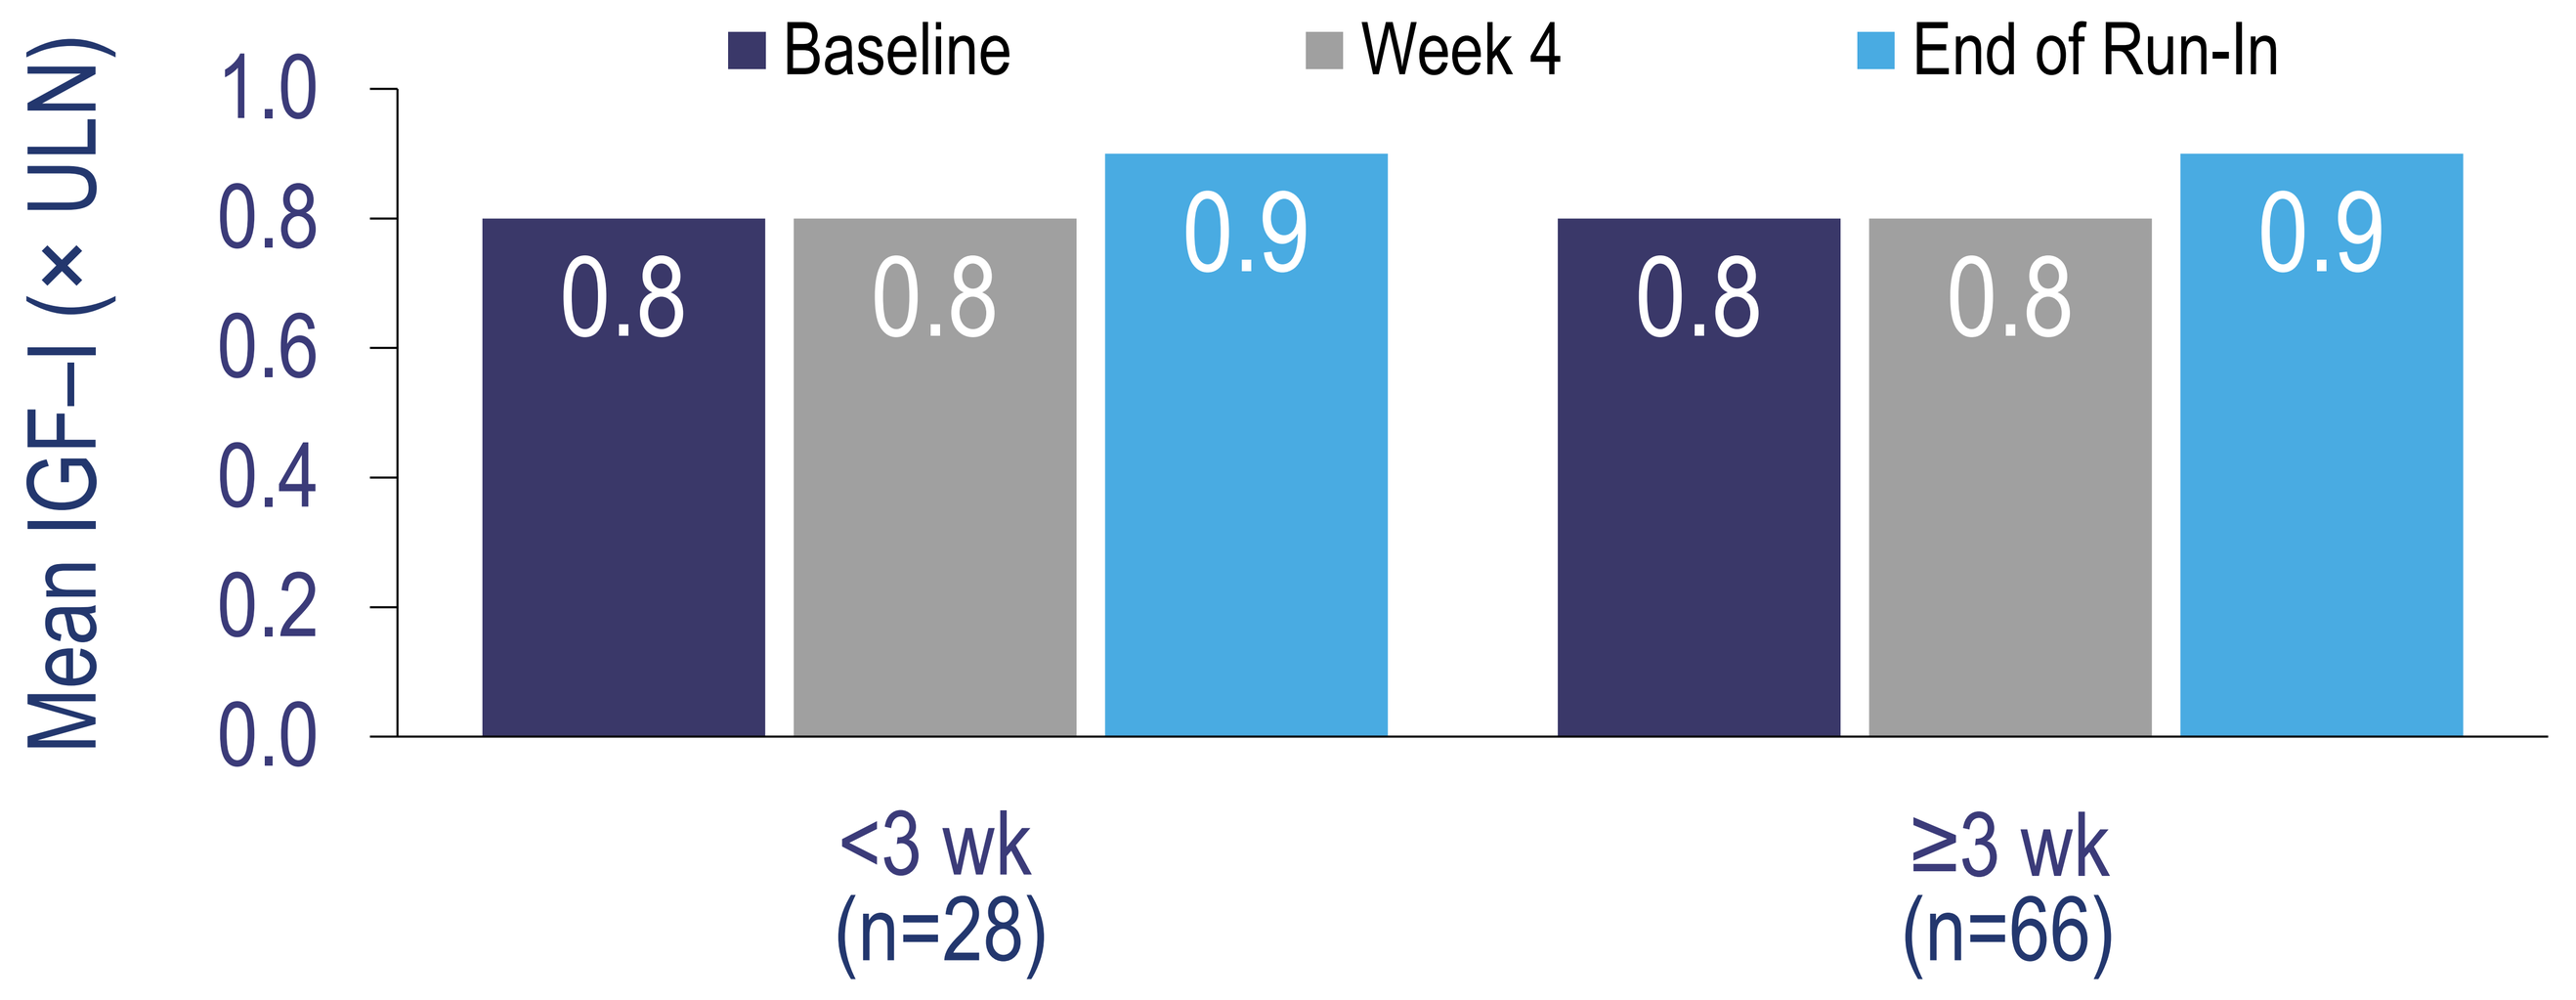
Baseline
Week 4
End of Run-In
Mean IGF–I (× ULN)
1.0
0.8
0.6
0.4
0.2
0.0
0.8
0.8
0.9
0.8
0.8
0.9
<3 wk
(n=28)
≥3 wk
(n=66)
Time since last iSRL dose

No differences were seen in acromegaly symptoms or patient-reported outcomes

- No significant differences in the Acromegaly Treatment Satisfaction Questionnaire (Acro-TSQ) domain scores were observed during the Run-in phase, regardless of OOC initiation timing
- Overall Acromegaly Index of Severity (AIS) scores decreased after the first month and at the end of the Run-in phase for both groups
 - –0.5 change from Baseline for the <3 weeks group and –0.4 for the ≥3 weeks group after the first month
 - –0.7 change from Baseline for the <3 weeks group and –0.2 for the ≥3 weeks group at the end of the Run-in phase

No new or increased safety signals were identified with a shorter time to initiation of OOC

Time since last iSRL dose

	<3 wk (n=40)	≥3 wk (n=105)
Patients with any TEAE, n (%)	26 (65)	79 (75.2)
AEs by System Organ Class (≥10%), n (%)		
General disorders and administration site conditions	5 (12.5)	15 (14.3)
Infections and infestations	9 (22.5)	30 (28.6)
Musculoskeletal and connective tissue disorders	1 (2.5)	20 (19.1)
Nervous system disorders	6 (15)	20 (19.1)
Respiratory, thoracic and mediastinal disorders	5 (12.5)	3 (2.9)
GI disorders	17 (42.5)	54 (51.4)
Related GI AEs by preferred term, n (%)		
Abdominal pain	2 (5)	17 (16.2)
Diarrhea	5 (12.5)	13 (12.4)
Flatulence	3 (7.5)	9 (8.6)
Nausea	9 (22.5)	22 (21)

GI, gastrointestinal; TEAE, treatment-emergent adverse event.

Conclusions

In this post hoc analysis of MPOWERED data, earlier initiation of OOC relative to the last dose of iSRL was not associated with differences in biochemical response or quality-of-life measures in patients who previously tolerated and responded to SRLs

No new or increased safety signals were identified, including drug-related GI events, with a shorter time to initiation of OOC

Further investigation is warranted to validate these results, which may support flexibility in the timing of OOC initiation relative to the last iSRL dose

References

1. Fleseriu M, et al. *Pituitary.* 2021;24:1-13.
2. Samson S, et al. *J Clin Endocrinol Metab.* 2020;105(10):1-13.
3. Fleseriu M, et al. *Lancet Diabetes Endocrinol.* 2022;10:102-111.

This poster is sponsored by Amryt Pharma. PRECISIONscientia provided medical writing assistance. Dr. Macut was a principal investigator at one of the sites participating in this study.

20th Congress of the European Neuroendocrine Association; September 7–10, 2022; Lyon, France